Pre-Clinical Caries Testing of an Improved Anti-Tartar Dentifrice Formulation

A.M. Pfarrer*

Procter & Gamble Company, Cincinnati, OH, USA

1194

ABSTRACT

The purpose of this experiment was to compare the anticaries efficacy of a new dentifrice formulation containing an improved anti-tartar ingredient (sodium polyphosphate, i.e. "poly"-pyrophosphate) to that of a clinically proven tartar control formula. The profile test chosen was an *in vitro* pH cycling experiment (White DJ; *Caries Res*. 21: 126-140. 1987). The model made use of 4mm enamel disks containing 96 hour Carbopol lesions (White DJ; *Caries Res*. 21: 228-242. 1987). The treatment regimen consisted of 1 minute soaks of treatment groups in 10 ml of 33% (w/w) dentifrice/saliva slurries 4 times per day. In between treatments specimens were soaked in pooled saliva with agitation. In between the 2nd and 3rd treatments, specimens were soaked in demineralizing solution to simulate acid challenge. After 5 days of treatment, fluoride uptake was determined using the microdrill biopsy technique. Products compared were: a) USP Reference Toothpaste: 1100ppm F (NaF), silica; b) new anti-tartar dentifrice: 1100ppm F (NaF), 7% sodium polyphosphate, silica; and c) placebo: 0ppm F (NaF), silica Fluoride uptake results [μg/cm^2 ± SD] were: a) 32.35 ± 4.03; b) 28.31 ± 7.56; and c) 2.96 ± 0.70, with a=b>c (p£0.05 ANOVA). **This experiment demonstrates the new anti-tartar dentifrice formulation has anticaries efficacy "at least as good" as the clinically proven USP Reference Standard Toothpaste.**

PURPOSE

The purpose of this experiment was to evaluate the fluoridating efficiency of a new tartar control dentifrice formulation, relative to a clinically-proven tartar control formula.

BACKGROUND

In the abscence of well-controlled clinical trials researchers and regulatory agencies rely on validated testing methodologies to provide laboratory confirmation of anticaries activity. These methods have evolved into validated and well controlled screens for anticaries product activity and their correlation with published clinical results is sufficient to qualify new formulations for efficacy.

The focus of current research is in the development of dentifrices capable of providing multiple benefits - including therapeutic and cosmetic benefits. The addition of new additives and improved therapeutic and excipient ingredients must be carried out carefully - since some ingredients could in theory influence potential anticaries actions of fluoride.

MATERIALS AND METHODS

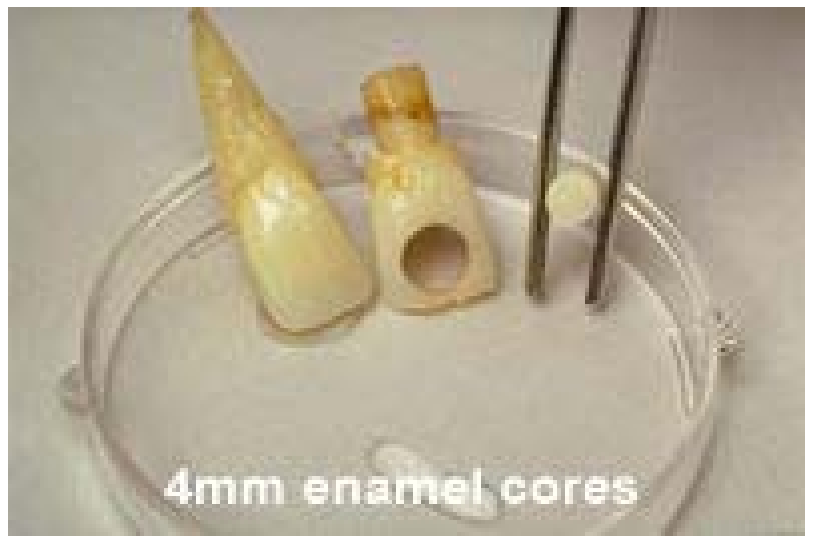
4mm enamel cores

Cores were mounted, ground and polished to remove approximately 50 micrometers of the natural surface

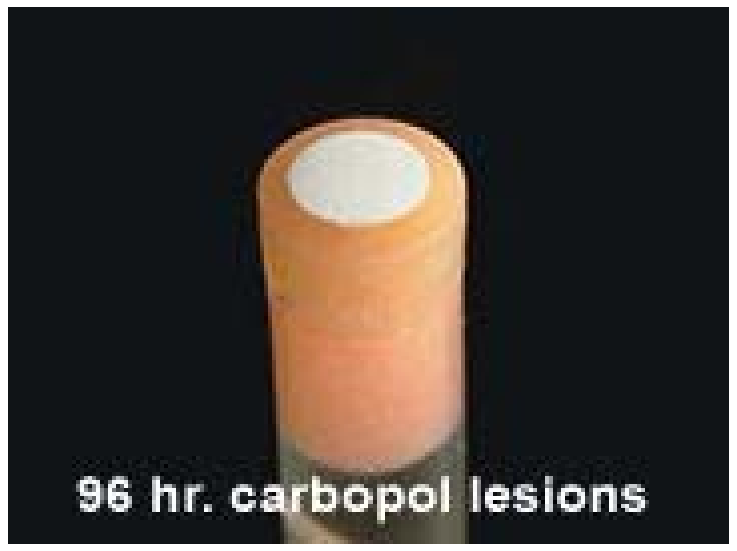
96 hr. carbopol lesions

Treatment Schedule

1 hr. saliva bath (initial pellicle formed)
1 min. treatment (1:3 slurry of dentifrice:saliva)
1 hr. saliva bath
1 min. treatment
1 hr. saliva bath
3 hr. demineralization (carbopol solution)
1 hr. saliva bath
1 min. treatmtent
1 hr. saliva bath
1 min. treatment
saliva bath overnight

Repeat cycle for 5 days

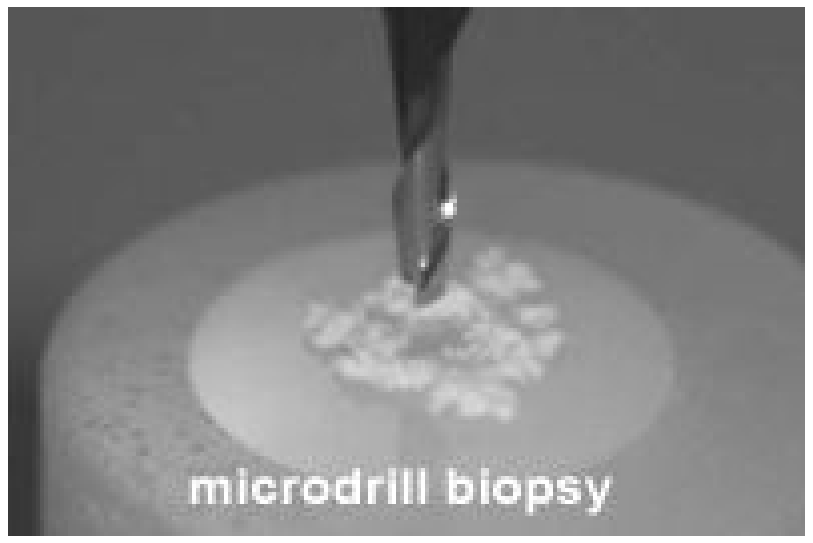
microdrill biopsy

RESULTS

Treatment	Fluoride Uptake (mg/cm^2)* (mean ± standard deviation)
Crest MultiCare Advanced Cleaning	**32.35 ± 4.03**
Crest Tartar Protection	**28.31 ± 7.56**
Placebo	**2.96 ± 0.70**

* Means within brackets are not significantly different (p≤0.05)

Research presented at the 78th General Session of the IADR, April 5-8, 2000