Laboratory Assessment of Tartar Control Efficacy of a Polyphosphate Dentifrice

E.R. Cox*, D.J. White
Procter & Gamble Co., Mason, OH, USA

1273

ABSTRACT

The efficacy of tartar control formulations can be assessed in the mPGM plaque biofilm calcification model (*J Dent Res* 71:173 (#543-544) 1992). The purpose of this experiment was to test activity of a 5.0 % polyphosphate (PP_n - *e.g.* "poly"pyrophosphate) dentifrice in comparison to clinically validated commercial antitartar formulations. Plaque biofilms were prepared and mineralized by alternate immersion of glass rods in human saliva and artificial mineralization solution supersaturated with respect to HAP, OCP, and DCPD (White et al., *J Dent Res.* 70; 276 (#84) 1991). Treatments of 25% w/w dentifrice water slurries were carried out for 30 seconds daily for 8 days, between saliva and mineralization solution immersions. Plaque calcium levels were determined by digestion and atomic absorbance spectroscopy. Test dentifrices included: A.) Crest® Regular NaF (control); B.) Colgate® Total (2% GantrezCo-Polymer); C.) Crest Tartar Control (5.0% Pyrophosphate); D.) Sanogyl® (0.7% Zinc Sulphate); and E.) Blend-a-Med® Synergy (5.0% PP_n). Results of the study (expressed as % reduction in mineralization vs. control producing 1035 mg calcium uptake) - a b students paired t: A.) 0.0, a; B.) 25.0, b; C.) 34.9, c; D.) 41.1, c; E.) 61.1, d. **Polyphosphate dentifrice produced plaque biofilm mineralization inhibition superior to pyrophosphate, co-polymer and zinc based tartar control dentifrices. These results support clinical testing of polyphosphate dentifrices as potential tartar control improvements.**

OBJECTIVE

The modified Plaque Growth and Mineralization Model (mPGM) is a technique where plaque is grown from human saliva under conditions to promote its mineralization. In our laboratories, this technique is used to:

1. define the tartar efficacy of a compound(s) in profile test applications;
2. examine kinetics of inhibitor reactions, and
3. assist in the determination of the mechanism of action of actives/formulations for tartar control.

The excellent correlation of clinical observations with mPGM test results allows the use of this technical profile test system for support of tartar control activity of products containing polyphosphate tartar agents in various dentifrice bases. The objective of this study was to test the tartar control activity of a polyphosphate, NaF dentifrice versus clinically proven control dentifrices.

MATERIALS AND METHODS

mPGM Plaque Biofilm Calcification Model:

1. Polished glass rods are used as the substrate
2. Saliva is collected daily by paraffin stimulation and pooled
3. Rods are dipped at 2 rpm for a ten day period in mineralizing solution (daily) and pooled saliva (overnight)
4. Rods are treated 1x per day in 25% wt./wt. dentifrice slurry
5. At conclusion of test, plaque biofilm is digested in acid and measured for calcium content via atomic adsorption.

Test Schematic:

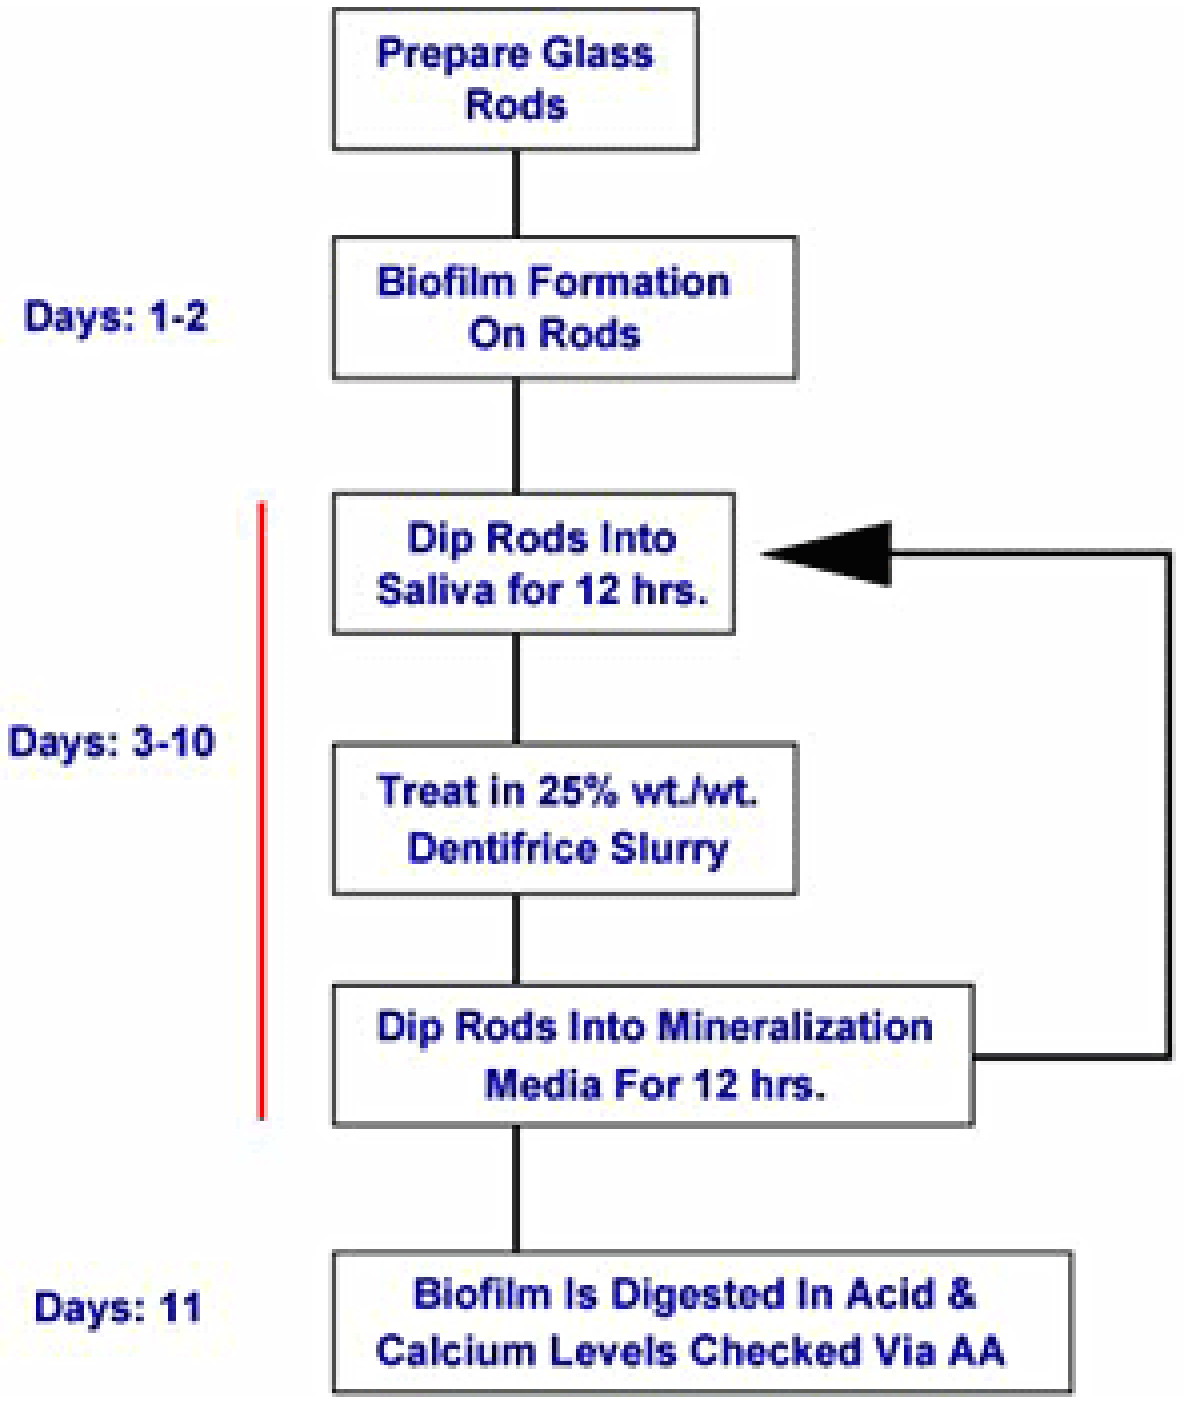
Prepare Glass Rods
Days: 1-2
Biofilm Formation On Rods
Dip Rods Into Saliva for 12 hrs.
Days: 3-10
Treat in 25% wt./wt. Dentifrice Slurry
Dip Rods Into Mineralization Media For 12 hrs.
Days: 11
Biofilm Is Digested In Acid & Calcium Levels Checked Via AA

RESULTS

mPGM Calcification Model

Product	% Red. vs. Control	Stat. Group
Crest Regular	na	a
Colgate Total	25.0%	b
Crest TC	34.9%	c
Sanogyl	41.1%	c
Blend-a-Med Synergy	61.1%	d
a¹b students paired t		

CONCLUSION

The results of the profile testing support significant clinical tartar control efficacy to be anticipated for a 5.0% polyphosphate, NaF dentifrice in comparison to clinically validated commercial formulations.

P&G Health Care Research Center

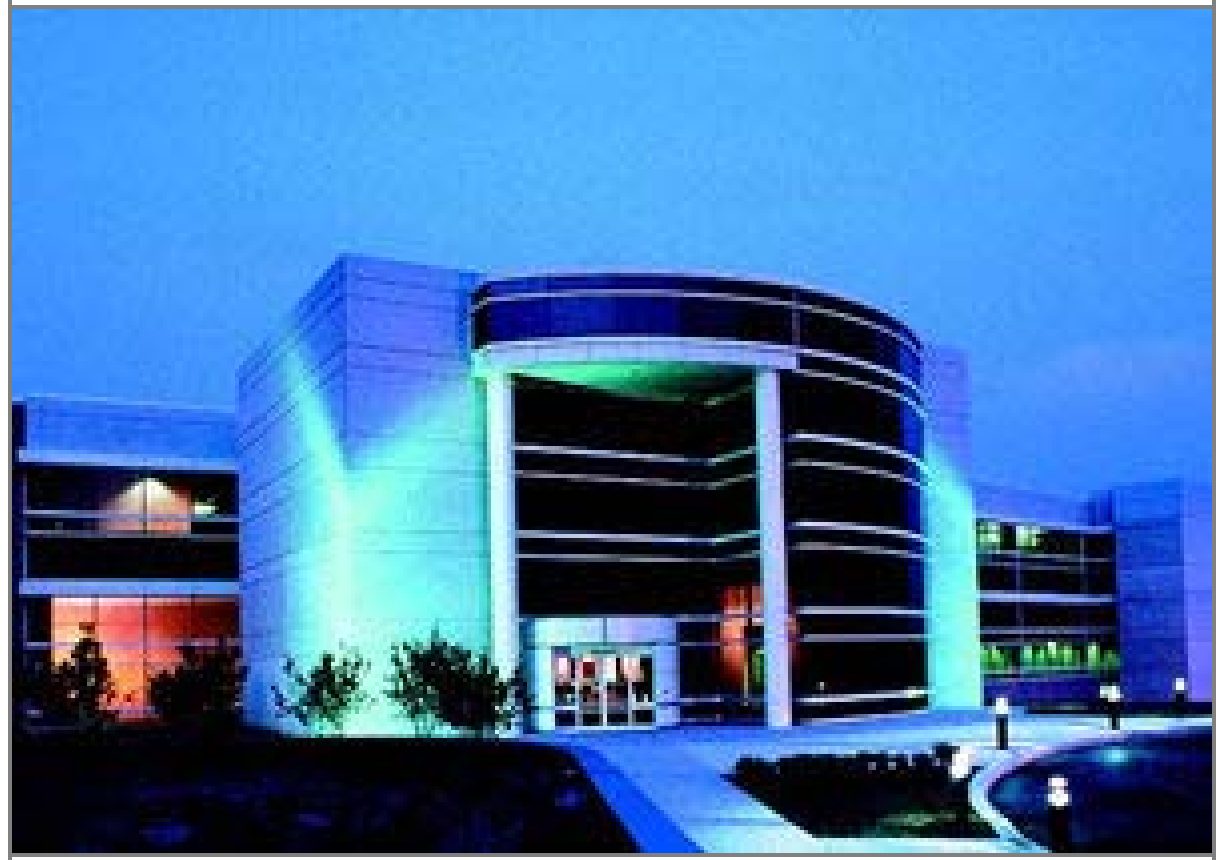

Mason, Ohio, USA

Research presented at the 78th General Session of the IADR, April 5-8, 2000

P&G