7-Day Partial Brushing Model Evaluating Gingival Epithelium Cytokine Levels

L.A. Bacca*, A.R. Biesbrock, W.M. Glandorf, M.S. Leusch
Procter & Gamble Co., Cincinnati, OH, USA

1302

ABSTRACT

Research has identified various gingival cytokines in crevicular fluid (GCF) as periodontial markers (Newman, et al, 1996). Prior work has shown that a dermal sampling tape (Biesbrock, et al, 1999) can be used to sample gingival epithelium and obtain measurable amounts of IL-1a and IL-1b. The objective of this pilot cross-over study was to evaluate if differences in certain cytokines (IL-1a/IL-1b/IL-6) are detectable in a 7-day partial-brushing model. Products tested were a marketed gingivitis dentifrice (Crest Gum Care, 0.454% SnF_2, CGC) vs. a control dentifrice (Advanced Formula Crest, 0.243% NaF, AFC). Gingival epithelium were sampled at t=0, 2, 4, and 7 days of partial brushing. Subjects brushed only the lingual dentition surfaces 2x/day with a 10 day wash-out period. Day 7 results: an increase from baseline in avg. IL-1a levels (pg/ml) was observed for the AFC leg (t=0: 2.06± 7.03;t=7:30.25±28.8) while avg. levels for CGC remained fairly constant (t=0: 8.57±5.46;t=7: 8.78±11.16); avg. AFC IL-1b levels (pg/ml) also increased (t=0: 2.99±2.78;t=7: 21.46±8.16), with a lower increase for the CGC leg (t=0: 4.31±1.27;t=7:12.76±16.84); avg. AFC IL-6 levels (pg/ml) again increased (t=0: 0.32±0.05;t=7:1.20±0.20), while the CGC avg. remained the same (t=0:0.30±0.03;t=7: 0.29±0.02). Upon returning to normal brushing, baseline levels of all three returned. **Results show differences in IL-1a, IL-1b, and IL-6 levels are detectable in this 7-day partial-brushing study, and a clinically proven gingivitis product maintained lower levels of these cytokine vs. the control dentifrice.**

PURPOSE

The objective of this study was to determine if a 7-day partial-mouth brushing study was a viable tool for evaluating: 1) whether certain cytokines are detectable after treatment with a dentifrices; 2) if the method is sensitive enough to differentiate between commercially available dentifrices.

BACKGROUND

There is an ongoing effort in the dental research community to identify potential biological markers of the early stages of gum disease, providing a valuable screening tool to dentists in identifying individuals at risk of severe gingival problems. Much of the periodontal research has centered on the evaluation of IL-1a, IL-1b, and TNF-a in GCF as potential diagnostic markers for periodontitis. Previous work by Biesbrock, *et al*, demonstrated that a dermal sampling adhesive strip (Sebutape) can be used to sample gingival tissue for the analysis of certain cytokines including IL-1a and IL-1b utilizing ELISA techniques. This work demonstrated a strong positive correlation between levels of IL-1α and IL-1β and clinical gingival inflammation. To date, these studies have not evaluated the effects of oral care products on cytokine levels in a non-brushing study. To avoid disrupting the dental plaque on the tooth surface, a partial-mouth brushing model was explored.

MATERIALS AND METHODS

Materials
Sebutape (CuDerm Corportation)
Quantikine IL-1a Assay Kit (R&D Systems)
Quantikine IL-1b Assay Kit (R&D Systems)
Quantikine IL-6 Assay Kit (R&D Systems)
Dulbecco's Phosphate Buffered Saline (Gibco)
Softmax ELISA Plate Reader and Software
Advanced Formula Crest (AFC) - 0.243% sodium fluoride
Crest Gum Care (CGC) - 0.454% stannous fluoride

Study Design
Number of Subjects: 5
Sebutape sampling time points: 0, 2, 4, 7 days of partial-brushing, and 5 days after returning to normal brushing with control dentifrice
Washout period: 10 days between treatments
Sampling site: gingival tissue from maxillary posterior right quadrant
Sampling Technique: gingiva dried with sterile gauze, Sebutape placed for 2 min.; sample is placed into sterile vial, sealed and frozen at -70 deg. C until analysis
Partial Brushing Technique: brush lingual surfaces only for at least 1 minute 2x/day

Sample Preparation/Analysis
Samples were allowed to equilibrate to room temperature. To each sample, 1 ml of Dulbecco's phosphate buffered saline was added. Samples were vortexed for 5 minutes. Cytokine levels of each sample were analyzed utilizing the procedure as described in the R&D Systems Quantikine Analysis Kits. Wash buffers and standards for all three cytokines were prepared the day prior to analysis and stored at 4 deg. C in sealed containers until needed.

Standards
IL-1a : standard curve range from 250 to 3.9 pg/ml
IL-1b : standard curve range from 250 to 3.9 pg/ml
IL-6 : standard curve range from 10 to 0.156 pg/ml

Data Analyses
A standard curve was constructed by doing a linear curve fit regression analysis vs. concentration of standard for IL-1a and IL-1b. As suggested by the R&D Quantikine kit, a linear curve was constructed for the IL-6 standards by plotting a log (concentration) vs. log (absorbance) to provide a more accurate curve fit. Sample concentrations were calculated from each curve generated. All standard curves and sample concentrations were generated by the program software for the Softmax ELISA Plate Reader.

RESULTS

In this partial-brushing study, measurable amounts of all three cytokines were observed. There was an increase in IL-1α, IL-1β, and IL-6 levels in the tissue samples as inflammation increased for the AFC treatment. CGC exhibited smaller increases in IL-1α and IL-1β levels with IL-6 levels remaining the same over the course of the study. There was a clear difference in the effects on the cytokines by the two different treatment groups with CGC maintaining lower levels of all 3 cytokines over AFC. Upon returning to normal brushing, levels of these cytokines returned to baseline levels. Results are as follows:

Research presented at the 78th General Session of the IADR, April 5-8, 2000

P&G

1302

7-Day Partial Brushing Model Evaluating Gingival Epithelium Cytokine Levels

L.A. Bacca*, A.R. Biesbrock, W.M. Glandorf, M.S. Leusch
Procter & Gamble Co., Cincinnati, OH, USA

Pg. 2

Treatment	Cytokine	Time pt.	level (std)
AFC	IL-1a	baseline	2.06 (7.03)
		day 7	30.25 (28.80)
	IL-1b	baseline	2.99 (2.78)
		day 7	21.46 (8.20)
	IL-6	baseline	0.32 (0.05)
		day 7	1.20 (0.20)
CGC	IL-1a	baseline	8.57 (5.46)
		day 7	8.78 (11.16)
	IL-1b	baseline	4.31 (1.27)
		day 7	12.76 (16.84)
	IL-6	baseline	0.30 (0.03)
		day 7	1.20 (0.20)

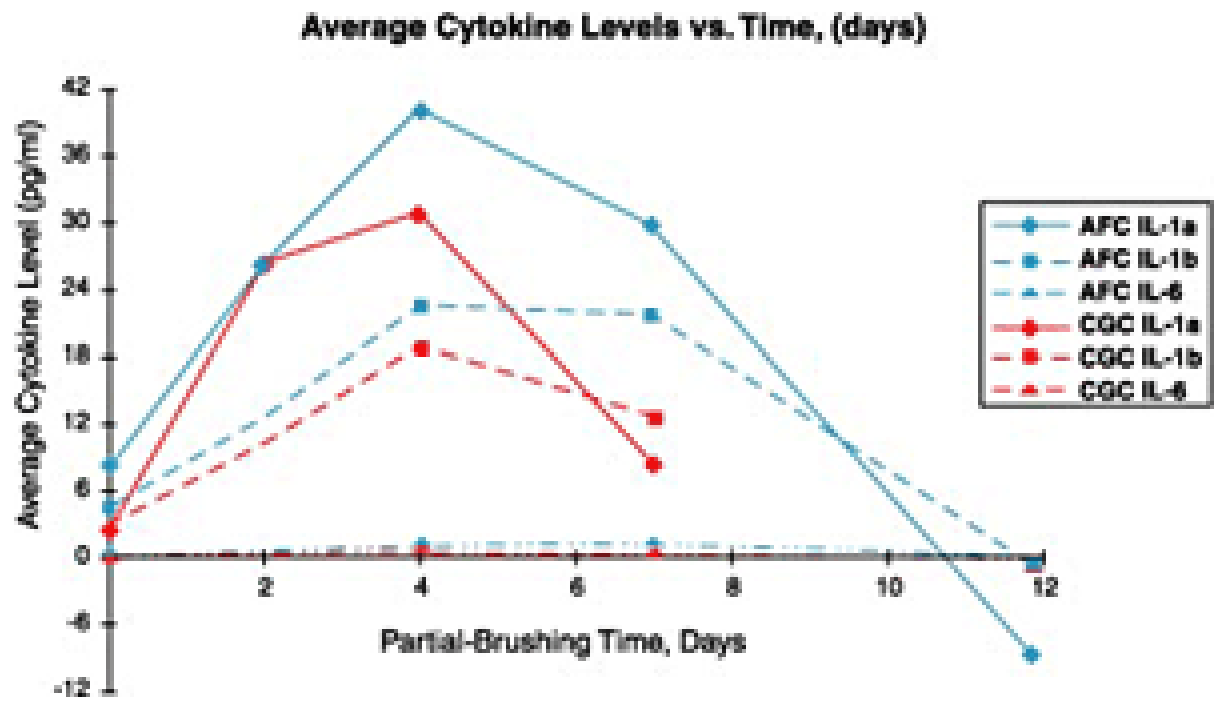
Average Cytokine Levels vs. Time, (days)
Average Cytokine Level (pg/ml)
42
36
30
24
18
12
6
0
-6
-12
2
4
6
8
10
12
Partial-Brushing Time, Days
AFC IL-1a
AFC IL-1b
AFC IL-6
CGC IL-1a
CGC IL-1b
CGC IL-6

CONCLUSION

Results from this study demonstrate that differences between IL-1α, IL-1β, and IL-6 levels are detectable in a 7-day partial-brushing study. In addition, a clinically proven gingivitis product (Crest Gum Care) maintained lower levels of these cytokines vs. the control (Advanced Formula Crest).

Research presented at the 78th General Session of the IADR, April 5-8, 2000