Novel *in vitro* Methodology Evaluating Potential Mucosal Irritation of Oral Care Formulations

1189

L.A. Bacca*, E.A. Jewell-Motz

P&G, Mason, OH, USA

ABSTRACT

Background: Oral irritation/tissue desquamation can result from exposure of the soft tissue to some dentifrice/rinse formulations. The potentially corrosive effects of common excipients, such as SLS, are well documented. As manufacturers develop products utilizing new actives or combinations of excipients and actives, testing methodology is needed to evaluate the irritation potential of these formulations and ingredients prior to human exposure. **Objective:** To develop *in vitro* methodology for the determination of potential oral irritation of oral care excipients/ formulations. **Methods:** *in vitro* tissue models (Mattek Corp.) of human oral keratinocytes were exposed to varying concentrations of common oral care excipients and dentifrice supernatants. These include SLS, poloxamer, NaF dentifrice, and pyrophosphate(PPi)/NaF dentifrices. As a measure of mucosal corrosivity, cell viability levels were monitored over time (up to 24 hrs) via the MTT assay. ET-50 values were calculated. IL1-α and IL1-β profiles were also monitored via ELISA. Correlations were made to *in vivo* human desquamation testing. **Results:** SLS solutions (0.2% to 0.6%) provided a clear dose response in both cell viability and cytokine levels. ET-50 values ranged from 10.5 to < 2 hrs. A dose response was observed for expressed levels of IL1-α and IL1-β. Addition of poloxamer to SLS solutions increased cell viability levels to the range of >24 to 9 hrs, and decreased levels of expressed cytokines. Treatments of NaF and PPi/NaF dentifrices (10% supernatants) exhibited differences for both ET-50 values and cytokine expression levels. These results are consistent and correlate with *in vivo* oral desquamation testing. Additionally, cell viability results provide a strong correlation to cytokine expression levels. **Conclusions: The *in vitro* tissue model provides a quick, reproducible method for evaluating the irritation potential of oral care excipients and prototype formulations. The methodology correlates well to human irritation results, and can provide a reliable alternative to animal testing.**

INTRODUCTION

Oral care formulations continue to evolve and provide new or multiple benefits such as whitening, cavity protection, tartar protection, gum health, etc. As these new formulations emerge, they are becoming more complex and contain more active ingredients and excipients. These formulations and excipients have the potential to be more harsh and pose a greater risk for causing some form of oral irritation in at least a portion of the population. Sodium lauryl sulfate (SLS) is a widely used surfactant in the dentifrice industry and has been implicated in oral mucosal irritation caused by some dentifrices. One of the most commonly occurring form of oral irritation is tissue desquamation resulting from exposure of the soft tissue to some dentifrice/rinse formulations. Currently oral irritation testing is done *in vivo* and can require several weeks to complete. A quick reproducible testing methodology is needed to evaluate the irritation potential of these formulations and ingredients prior to human exposure. In the skin care industry, *in vitro* tissue culture models are routinely used to evaluate the potential irritation associated with product usage. These methods utilize cell viability and cytokine expression analyses, specifically IL-1α and IL-1β, as measures of a products potential corrosivity. Skin researchers have investigated IL-1α and IL-1β because of their early involvement in the inflammatory cascade with elevated levels of these cytokines being observed in inflamed tissue. Recent advances have made similar tissue cultures systems available that are constructed from human buccal epithelial cells. This tissue model may allow for *in vitro* oral irritation/desquamation testing of various new excipients and/or formulations prior to human exposure.

PURPOSE

The purpose of this investigation was to develop an *in vitro* testing methodology utilizing human buccal tissue cultures that would predict the oral irritation potential of potential oral care excipients and formulations prior to human exposure.

MATERIALS AND METHODS

Materials:

Human Buccal Epithelial Tissue Culture System

- EpiOral™ tissue culture system - three-dimensional model of noncornified human buccal epithelium (MatTek Corp., ORL-100)
- A three-dimensional model of human buccal epithelium 8-12 layers thick

MTT Assay kit (MatTek Corp.)

- a quantitative colorimetric assay of cell proliferation
- measures the reduction of a yellow tetrazolium product into a purple formazan product by the mitochondria of viable cells

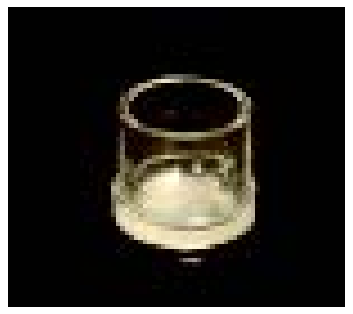

Fluorokine MAP Multiplex Cytokine Assay Kits (R&D Systems)

- liquid suspension of microscopic beads with a unique spectral address for each analyte
- analytes evaluated include IL-1α and IL-1β

Treatments

1) water
2) 10% (w/w) supernatant of a 0.243% NaF dentifrice
3) 10% (w/w) supernatant of a 0.243% NaF/5% PPi dentifrice
4) 0.2% (w/w) aqueous SLS
5) 0.4% (w/w) aqueous SLS
6) 0.6% (w/w) aqueous SLS
7) 0.2% (w/w) aqueous SLS w/0.2% poloxamer 407
8) 0.4% (w/w) aqueous SLS w/0.2% poloxamer 407
9) 0.6% (w/w) aqueous SLS w/0.2% poloxamer 407

Methods:

Cell culture treatments

- Cell culture inserts are incubated overnight at 37°C and 5% CO_2. On treatment day, inserts are moved to a 6-well plate containing appropriate culture medium. Each insert is treated with 100 ìl of freshly prepared treatment solution and returned to incubation. Media samples and cell culture inserts are pulled at baseline, 2, 6, and 24 hrs for cytokine expression profile analyses and MTT assays. Treatments for each time point and each treatment are performed in triplicate

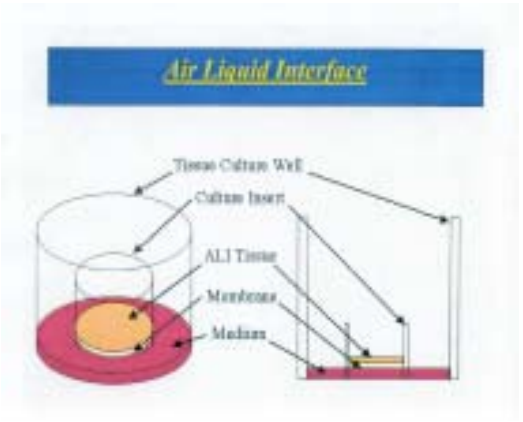
Air Liquid Interface

MTT Assay

- Following treatment, cell culture rafts are washed with PBS solution and placed into a yellow tetrazolium dye solution (MTT, 1mg/ml) and incubated at 37°C and 5% CO_2 for 3 hrs. The insoluble purple formazin product is then extracted with isopropyl alcohol overnight at room temperature in the dark. A 200 ìl aliquot is pulled and absorbances read at 570 nm. Percent viability is calculated versus the water control. A plot is constructed using a semi-log scale of % viability vs. dosing time and the time of exposure which reduced viability to 50% (ET-50) is calculated.

Cytokine Expression Analyses

- Cell culture media were analyzed for IL-1α and IL-1β expression profiles utilizing the Luminex xMAP technology. A sample aliquot is added to a 96-well plate containing microscopic beads pre-coated with analyte-specific antibodies. Following incubation and addition of conjugate solution, cytokine levels are measured with the Bio-Rad Bio-Plex™ system.

RESULTS

SLS Treatment Effects

A clear dose-response was observed for SLS for both cell viability and cytokine expression levels. As levels of SLS increased, cell viability decreased. ET-50 values ranged from 10.5 hrs to < 2hrs. The addition of poloxamer decreased the toxicity of the SLS solutions. ET-50 values significantly increased in range. Levels of IL-1α and IL-1β increased with SLS concentration. The addition of poloxamer decreased observed cytokine levels at the corresponding SLS concentrations while exhibiting a dose-response. [1]

Dentifrice Treatment Effects

The marketed dentifrice supernatants exhibited similar profiles for cell viability and cytokine expression. Both dentifrices demonstrated ET-50 values > 24 hrs. There were however slight differences in the profiles of viability and cytokine expression. The NaF/PPi dentifrice produced a different cell toxicity profile with slightly lower viability levels. IL-1α and IL-1β expression levels were also slightly higher than the NaF dentifrice.

Treatment	Estimated ET50 (hrs)	Mean IL -1α ± std (pg/ml)	Mean IL -1β ± std (pg/ml)
Water	na	60.6 ± 6.7	63.34 ± 18.7
0.2% SLS	10.5	275.5 ± 26.2	326.9 ± 31.1
0.4% SLS	4.2	365.6 ± 4.6	451.8 ± 28.6
0.6% SLS	<2	432.8 ± 21.1	566.2 ± 29.2
0.2% SLS/0.2% poloxamer	>24	132.9 ± 5.7	177.3 ± 22.3
0.4% SLS/0.2% poloxamer	9.8	196.7 ± 7.1	269.3 ± 14.7
0.6% SLS/0.2% poloxamer	8.9	206.9 ± 16.6	333.9 ± 19.9
10% NaF dentifrice	>24	162.45 ± 28.3	223.13 ± 22.8
10% NaF/PPI dentifrice	>24	216.32 ± 32.0	223.13 ± 22.8

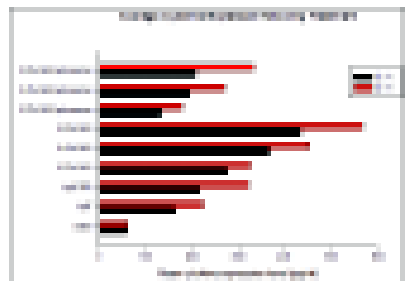

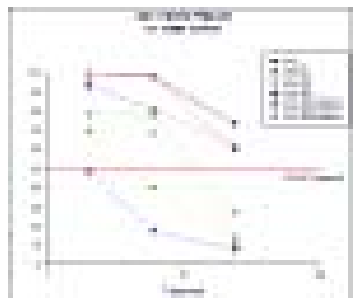

Overall results

A strong correlation is observed between the cytokine expression profile and the MTT cell viability results (r^2=0.88 to 0.92). As cytokine levels increase, cell viability decreases. The increase in cytokine expression and decrease in cell viability observed for the SLS treatments is consistent with the literature showing SLS toxicity to oral mucosal tissue. The addition of poloxamer to SLS solutions demonstrated a decrease in toxicity with cell viability increasing and cytokine profiles decreasing which is also consistent with manufacturers data. The slight increase in cytokine expression and decrease in cell viability observed for the NaF/PPi dentifrice is consistent with *in vivo* human oral irritation testing where PPi containing dentifrices may cause slight irritation for select individuals. The results of these studies were consistent across multiple experiments with differing lots of cell culture tissue inserts. In addition, experiments utilizing this methodology demonstrated low cell viability and high cytokine expression levels for experimental dentifrice formulations that failed in vivo desquamation testing.

CONCLUSION

- The *in vitro* tissue model provides a quick, reproducible method for evaluating the irritation potential of oral care excipients and prototype formulations. The methodology correlates well to human irritation results, and can provide a reliable alternative to animal testing.

MARCH 9-12, 2005

P&G

Research presented at the 83rd General Session of the IADR, March 9-12, 2005