Comparative Effects of Cetylpyridinium Chloride Mouthrinses on Plaque Glycolysis and Regrowth

L.A. Bacca, M.J. Doyle*, D.M. Macksood, A.C. Lanzalaco

Procter & Gamble Co., Mason, OH, USA

2749

ABSTRACT

Mouthrinses containing CPC have been shown to be effective in the control of dental plaque (Renton-Harper, et al, *J. Perio*., 67:486-489, 1996). CPC is a quaternary ammonium compound, cationic in nature, which allows the compound to exhibit powerful antimicrobial activity. The compound's availability and corresponding biological activity can be hindered by other ingredients in mouthrinse formulations (Addy, et al, *J. Dent. Res*., 72:719, 1993). The objective of the present study was to determine, utilizing the plaque glycolysis and regrowth model (PGRM), whether variations in rinse formulations effect CPC biological activity. This study evaluated both commercially available CPC rinses and experimental CPC rinses optimized for CPC availability. The following mouthrinses were evaluated: 1.)Scope® (SCP: Procter & Gamble Co., 0.045% CPC); 2.)Cepacol® (CEP: J.B. Williams, 0.05% CPC); 3.)Act® (Johnson & Johnson, 0.05% NaF/0.05% CPC); 4.)CP1 (placebo, 0% CPC); 5.) CP6 (water); and 6-9.)experimental CPC rinses containing 0.025% (CP2), 0.05% (CP3), 0.075% (CP4), and 0.1% (CP5) CPC. The study design utilized 9 subjects in a crossover fashion where each subject served as his/her own control. On the morning of the test, subjects presented to the site having refrained from oral hygiene the previous 12 hours. Baseline plaque samples were collected from the upper dentition, and the subjects rinsed with the assigned test formulation for 10 seconds. Additional plaque samples were taken from the lower left and lower right quadrants at 15 and 45 minutes post-brushing respectively. Normalized plaque biomasses, incubated in TSB buffer, were assayed for plaque glycolytic and regrowth activities using standard PGRM methodology (White, et al, *J. Clin. Dent.*, VI:59-70, 1995). Analysis of treatment effects by time (measure as area under the curve: AUC) revealed the following mean AUC results. For plaque glycolysis: 1.)SCP: 24.25b; 2.)CEP: 24.21b; 3.)ACT: 1.51a; 4.)CP1: -1.56a; 5.)CP2: 17.29b; 6.)CP3: 27.76b; 7.)CP4: 37.02c; 8.)CP5: 44.74c; 9.)CP6: -1.19a (c>b>a:t-test p<0.05). For regrowth: 1.)SCP: 184.62b; 2.)CEP: 176.4b; 3.)ACT: 42.88a; 4.)CP1: 21.16a; 5.)CP2: 116.49b; 6.) CP3: 198.8b; 7.)CP4: 194.78b; 8.)CP5: 206.53b; 9.)CP6: -20.48a (b>a:t-test p<0.05). **These results show that mouthrinses optimized for compatibility with CPC were significantly more effective at reducing plaque glycolytic and regrowth activity than the current commercially available rinses.**

INTRODUCTION

Cetylpyridinium chloride (CPC) mouthrinses have been shown to be effective in the control of dental plaque (Renton-Harper, et al, *J. Perio*., 67:486-489, 1996). This quaternary ammonium chloride compound's availability and corresponding biological activity has been shown to be hindered by other mouthrinse ingredients (Addy, et al, *J. Dent. Res*., 72:719, 1993). The Plaque Glycolysis and Regrowth Model (PGRM) is an *in situ* method that allows evaluation of a formulation's biological activity, based on its effects on plaque metabolism.

OBJECTIVE

The aim of the present study was to assess, using PGRM, to what degree variations in rinse formulations affected CPC's biological activity.

MATERIALS AND METHODS

Formulations Tested

1. Scope® (SCP) - 0.045% CPC (P&G)
2. Cepacol® (CEP) - 0.05% CPC (J.B. Williams)
3. Act® (ACT) - 0.05% CPC/0.05% NaF (Johnson & Johnson)
4. Placebo (CP1) - 0.0% CPC (P&G)
5. Experimental CPC (CP2) - 0.025% CPC (P&G)
6. Experimental CPC (CP3) - 0.05% CPC (P&G)
7. Experimental CPC (CP4) - 0.075% CPC (P&G)
8. Experimental CPC (CP5) - 0.1% CPC (P&G)
9. Water (CP6) - 0% CPC

Study Design

A crossover design was used in which 9 subjects rinsed with each of the 9 test products.

Test Product Application/Plaque Collection

Subjects presented to the site having refrained from oral hygiene the previous 12 hours. Subjects collected baseline plaque from the maxillary dentition with a sterile polyester swab. Subjects then rinsed for 10 seconds with the assigned test product. Treated plaque samples were collected by subjects from the lower left and right quadrants of the mandibular dentition 15 and 45 minutes post-treatment respectively.

Plaque Sample Preparation

Swabs containing plaque samples were placed in 1.75 ml of 0.03% BBL Trypticase Soy Broth and vortexed for 15 seconds. The resulting suspended plaque samples were then normalized for plaque biomass by adjusting to a constant optical density (OD) of 0.2 absorbance units with 0.03% TSB solution.

For Glycolysis/Regrowth

One ml of the normalized plaque/TSB solution was added to 50 ul of 40% sucrose solution in a 2ml Eppendorf vial. The plaque samples (pH=7.2) were then incubated for 2 hrs at 37C in an Eppendorf Thermomixer at 1200 rpm agitation. Acid production was determined by measuring the pH of the plaque samples following 2 hrs of incubation. A 300 ul sample of the normalized plaque/TSB solution was added to a 2 ml Eppendorf vial containing 0.5 ml of 6% (w/w) BBL TSB and 100 ul of sterile water. Bacterial growth was accelerated by the addition of 50 ul of a 40% sucrose solution. Prior to incubation, the OD of the plaque sample was measured at 600 nm. Samples were incubated for 4 hrs at 37C in an Eppendorf Thermomixer at 1200 rpm agitation. After incubation, the OD of the plaque samples was measured following homogenization with a pellet mixer. Regrowth was determined by calculating the ratio of the post-incubation OD to the pre-incubation OD.

Research presented at the 75th General Session of the IADR; Orlando, Florida March 19-23, 1997

P&G

2749

Comparative Effects of Cetylpyridinium Chloride Mouthrinses on Plaque Glycolysis and Regrowth

L.A. Bacca, M.J. Doyle*, D.M. Macksood, A.C. Lanzalaco
Procter & Gamble Co., Mason, OH, USA

Pg. 2

Calculation of Area Under the Curve

The post-incubation pH's and OD ratio's of the treated plaques of various sampling points were compared graphically to the final pH's and OD's of the baseline plaque samples. AUC's were calculated from the area between the baseline plaque pH and OD ratio and the treated plaque pH and OD ratio.

RESULTS

Treatment	Mean AUC Glycolysis*	Mean AUC Regrowth*
CP6	-1.19(2.92)a	-20.48(23.19)a
CP1	-1.56(7.17)a	21.16(105.6)a
ACT	1.51(4.77)a	42.88(76.53)a
CP2	17.29(13.9)b	116.49(89.74)b
CEP	24.21(19.36)b	176.40(115.34)b
SCP	24.25(14.89)b	184.62(167.57)b
CP3	27.76(22.88)b	198.8(147.21)b
CP4	37.02(24.24)c	194.78(129.44)b
CP5	4.74(12.36)c	206.53(152.06)b

c>b>a, $p \le 0.05$; t-test
*mean of 9 treatments/test formulation
Parentheses enclose standard deviations

Figure 1: Change in pH

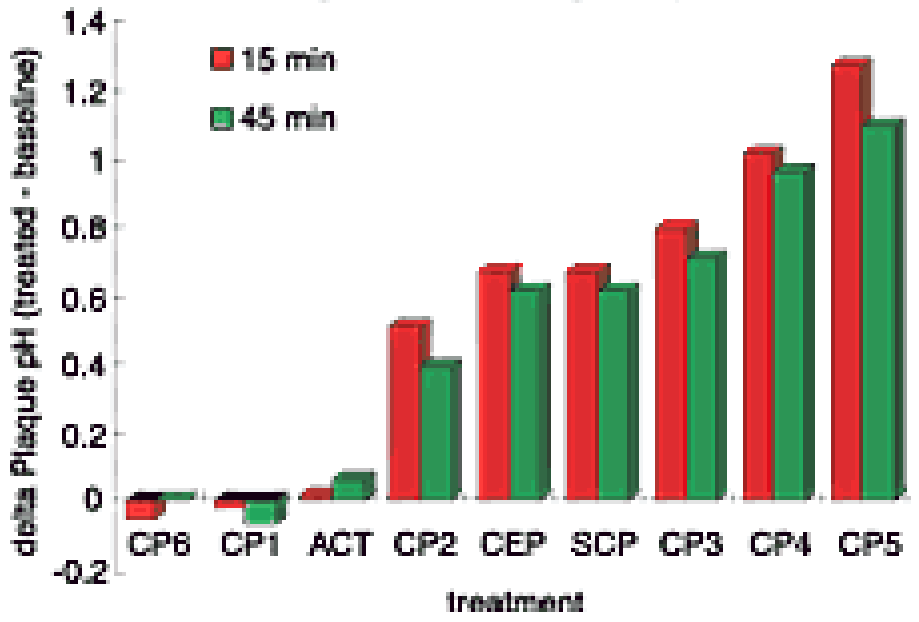
1.4
1.2
1
0.8
0.6
0.4
0.2
0
-0.2
15 min
45 min
delta Plaque pH (treated - baseline)
CP6
CP1
ACT
CP2
CEP
SCP
CP3
CP4
CP5
treatment

CONCLUSION

These results show that mouthrinses optimized for compatibility with CPC were significantly more effective at reducing plaque glycolytic and regrowth activity than some commercially available rinse products.

Research presented at the 75th General Session of the IADR; Orlando, Florida March 19-23, 1997

P&G