731

Development of a Non-Radiolabeled Solid-Phase Glucosyltransferase Assay

K.Herbort*, R.L. Wimalasena, E.A. Goulbourne Jr.
Procter & Gamble Co., Mason, OH, USA

ABSTRACT

Glucosyltransferase (GTF) from mutans streptococci in the oral cavity, breaks down sucrose to glucose and fructose and subsequently forms soluble and insoluble glucans as the end product. Glucans formed by GTF promote bacterial colonization of the tooth, and bacterial metabolism results in the production of acid which causes cavities. One of the strategies to prevent caries is to use potent inhibitors of the adsorbed GTF to prevent bacterial colonization. To determine the efficacy of potential GTF inhibitors, it was necessary to have a method to measure the activity of adsorbed enzyme. The method traditionally used employs radiolabeled sucrose and takes 24 hr. to obtain results. We have developed a faster (3 hr.) non-radioactive method with good reproducibility (less than 10% RSD) to measure the activity of adsorbed enzyme. Briefly, ceramic hydroxyapatite powder is added to a column and washed to remove particulates. GTF, which has a very high binding affinity for hydroxyapatite, is then added to the column and the column is washed again. Treatment is applied for 1 minute at room temperature, followed by several washes to remove excess active. A sucrose solution is added to the column, and the column is incubated for 30 minutes at 37 ^{o}C. The effluent is collected and assayed for glucose content, one of the breakdown products of sucrose by GTF. There was a direct correlation between the amount of glucan formed and the amount of free glucose measured. This method has increased the throughput of the screening work and for the first time eliminated the use of radioactive material. This assay was used to determine the inhibitory activity of experimental and commercial products. Results confirmed the expected behavior of known GTF inhibitors and provided information on the mechanism of action of experimental compounds.

INTRODUCTION

Streptococcus mutans has been implicated in the development of caries due to the ability of this organism to a.) convert sucrose to glucans, which aids in bacterial colonization of the tooth surface and b.) produce lactic acid through glycolysis of sugar. GTF produced by cariogenic bacteria is largely responsible for these activities by hydrolyzing sucrose to glucose and fructose, and forming glucans through the polymerization of glucose.

$$1.\ \textit{Sucrose} \xrightarrow{\textit{GTF}} \text{Glucose} + \textit{Fructose}$$

$$2.\ \text{Glucose} \xrightarrow{\textit{GTF}} \textit{Glucans}$$

$$3.\ \text{Glucose} \xrightarrow{\textit{Glycolysis}} \textit{Lactic acid}$$

The solid-phase GTF enzyme assay was developed to identify potential actives which cause an irreversible inhibition of GTF. Actives which are weakly bound to the solid support should disassociate and be washed from the column. On the other hand, those actives which are more tightly bound will continue to show their activity, even with excessive washes. GTF activity is determined by measuring glucose, an intermediate product of the enzyme reaction.

MATERIALS AND METHODS

Relationship Between Glucan Formation and Glucose Measurements

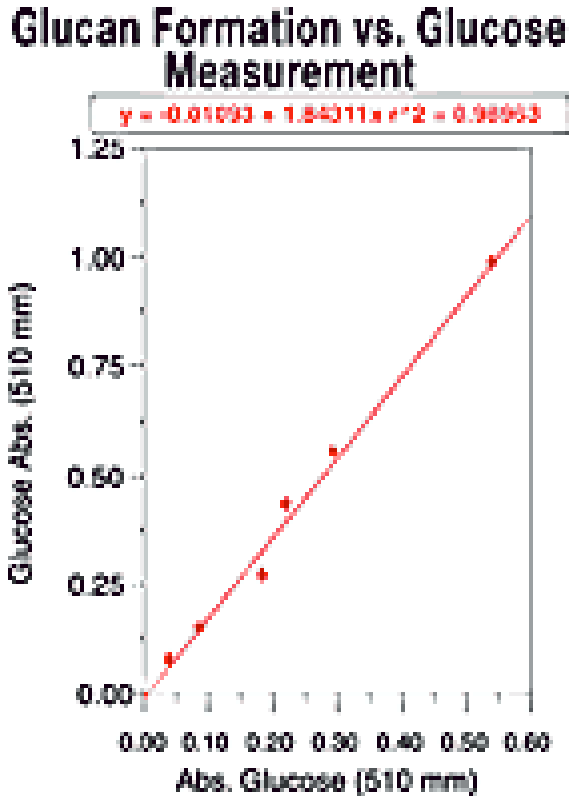
Glucan Formation vs. Glucose Measurement
Glucose Abs. (510 mm)
1.25
1.00
0.75
0.50
0.25
0.00
0.00 0.10 0.20 0.30 0.40 0.50 0.60
Abs. Glucose (510 mm)

Procedure:

1. Add different concentrations of GTF to separate tubes.
2. Add 600 mL 100 mM sucrose in 0.1M Acetate Buffer, pH 5.6.
3. Incubate 1 hr. at 37^{o} Celsius.
4. Measure glucan formation by recording absorbency of solutions at 550 nm.
5. Spin each solution down for 2 minutes and assay the supernatant for glucose content.

1. Add HAP

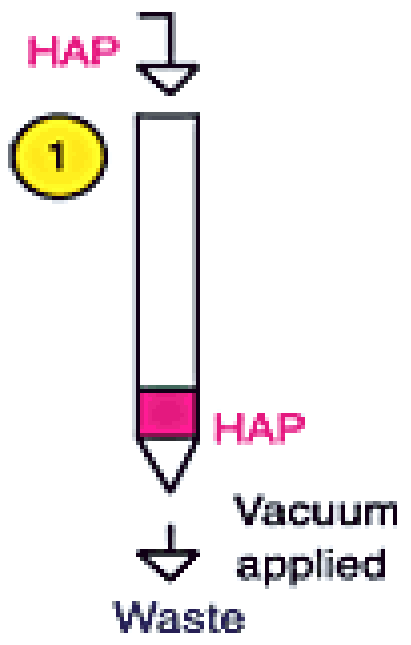
HAP
1
HAP
Vacuum applied
Waste

-Add 150 mg hydroxyapatite per column

-Wash: Apply vacuum and add 4 X 1ml filtered water

2. Add GTF

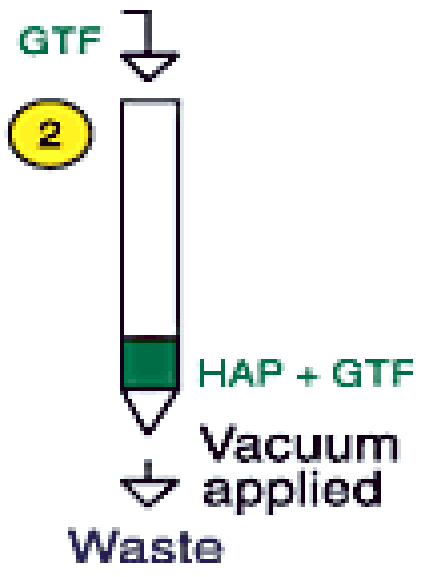
GTF
2
HAP + GTF
Vacuum applied
Waste

-Add GTF to each column

-Wash: Apply vacuum and add 2 X 3ml buffer

Research presented at the 75th General Session of the IADR; Orlando, Florida March 19-23, 1997

731

Development of a Non-Radiolabeled Solid-Phase Glucosyltransferase Assay

K.Herbort*, R.L. Wimalasena, E.A. Goulbourne Jr.
Procter & Gamble Co., Mason, OH, USA

Pg. 2

3A. Add Control

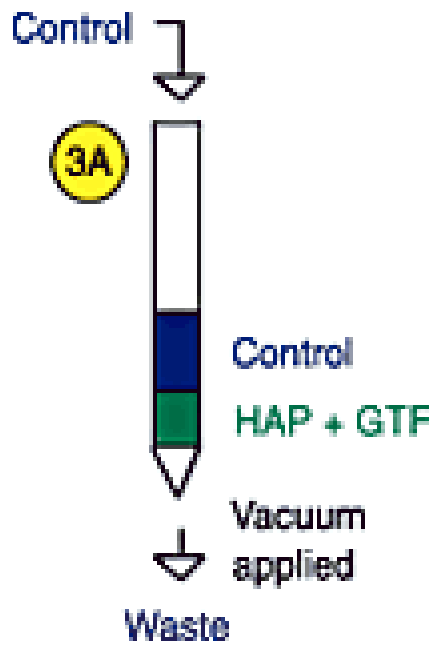
Control
3A
Control
HAP + GTF
Vacuum applied
Waste

-Add 0.5 ml control to the control columns

-Vortex briefly and incubate for 1 min. at RT

-Wash: Apply vacuum and add 8 X 3ml buffer

3B. Add Treatment

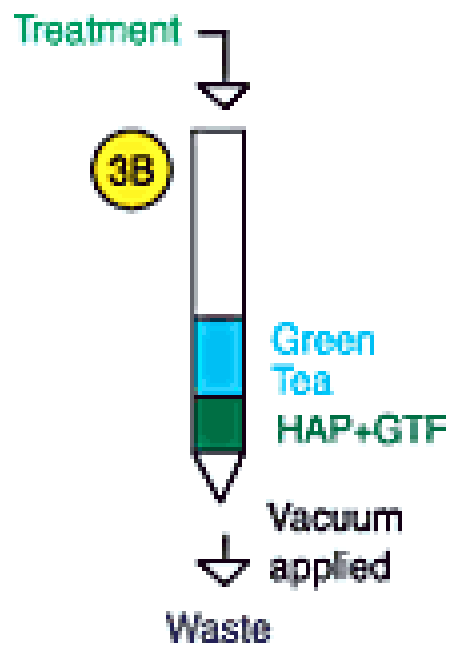
Treatment
3B
Green Tea
HAP+GTF
Vacuum applied
Waste

-Add 0.5 ml treatment to the treatment columns

-Vortex briefly and incubate for 1 min. at RT

-Wash: Apply vacuum and add 8 X 3ml buffer

4. Add Sucrose

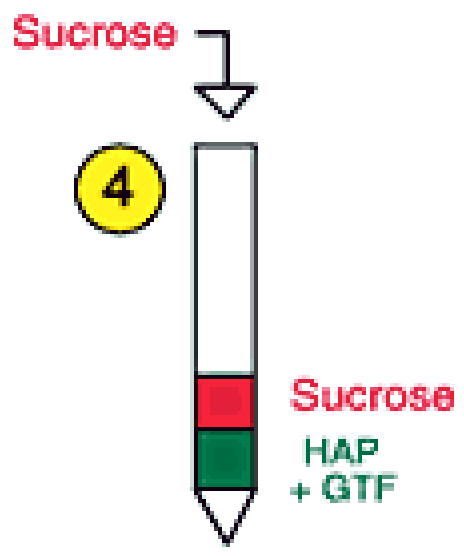
Sucrose
4
Sucrose
HAP + GTF

-Add 0.5 ml sucrose/column

-Vortex briefly and incubate for 30 min. at 37° C

-Mix columns every 10 min. during incubation

5. Collect effluent/Measure glucose

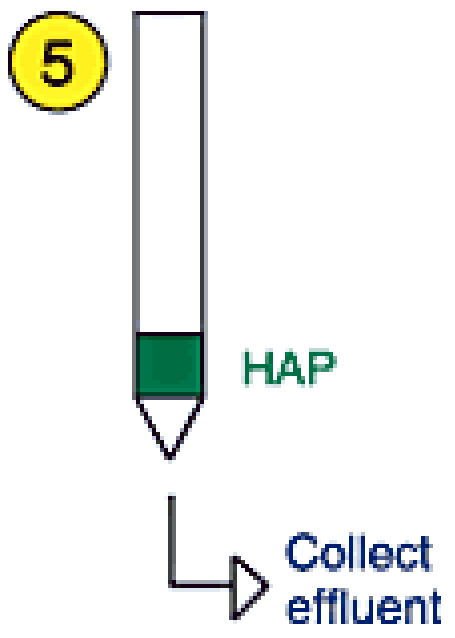
5
HAP
Collect effluent

-Apply vacuum and collect effluent

-Analyze 100 microliters effluent for glucose content

DETECTION METHOD FOR GLUCOSE MEASUREMENT
• Glucose measured using Glucose Trinder Reagent (Sigma Diagnostics) • Change in absorbency measured at 510 nm for 2 minutes • Calibration curve is linear from 0.025 mg/ml - 0.5 mg/ml • 100 mM fructose did not react with the Glucose Trinder Reagent

Assay Calibration

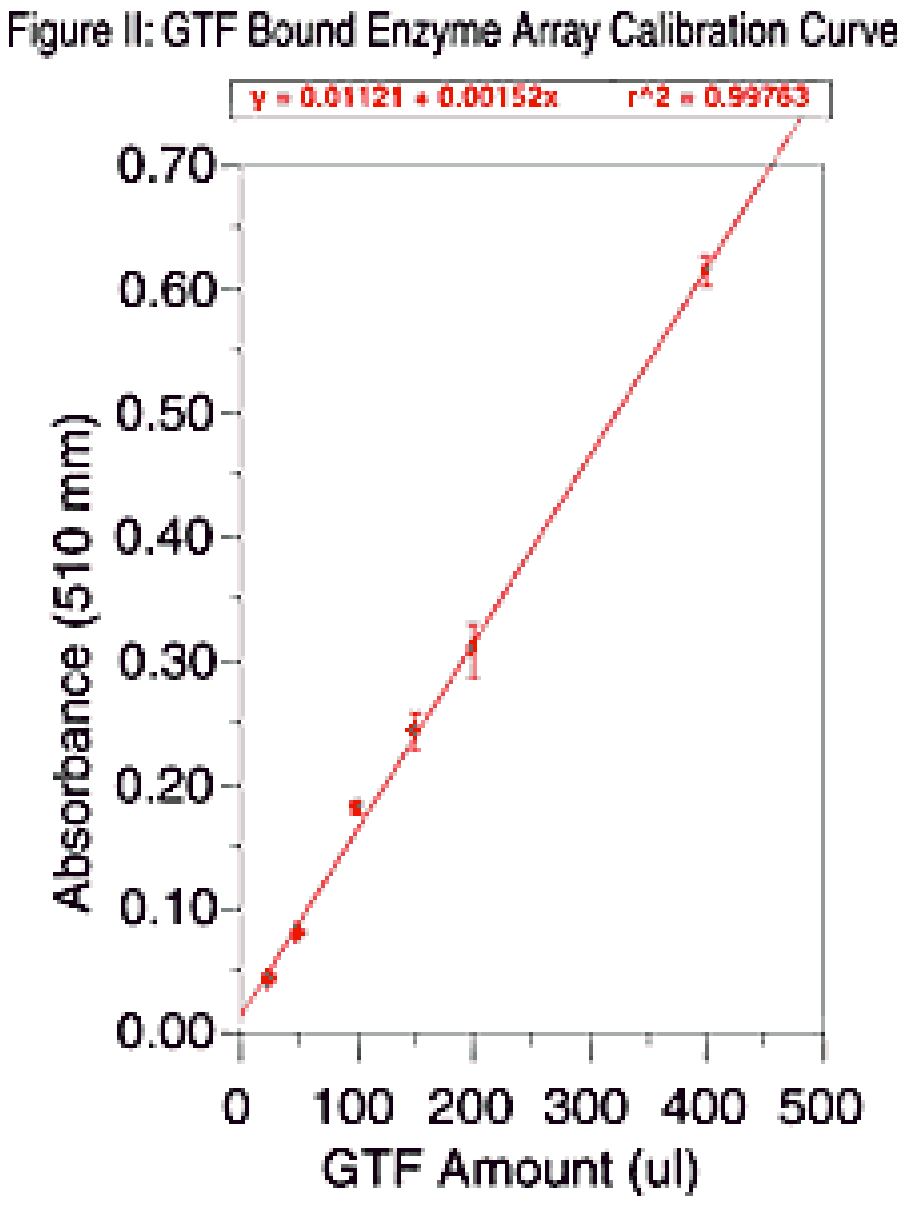
Figure II: GTF Bound Enzyme Array Calibration Curve
y = 0.01121 + 0.00152x r^2 = 0.99763
Absorbance (510 mm)
0.00
0.10
0.20
0.30
0.40
0.50
0.60
0.70
0
100
200
300
400
500
GTF Amount (ul)

Procedure:

The protocol described above was utilized with the exception that different amounts of GTF were added to the column and buffer was added during the treatment step, acting as a control.

Research presented at the 75th General Session of the IADR; Orlando, Florida March 19-23, 1997

731

Development of a Non-Radiolabeled Solid-Phase Glucosyltransferase Assay

K.Herbort*, R.L. Wimalasena, E.A. Goulbourne Jr.
Procter & Gamble Co., Mason, OH, USA

Pg. 3

RESULTS

Test compound	Percent Reduction GTF Activity
Green tea extract (approx. 0.3-0.5% polyphenols)	70
1% oolong tea (approx. 0.5% polyphenols)	50
Epicatechin gallate (1 mg/ml)	none
Epigallocatechin gallate (1 mg/ml)	none
500 ppm Copper glycinate	20
Peridex	none
Listerine	none
N-methyl deoxynojiromycin (5 mg/ml)	none
5% cranberry extract	50
10% fructose	none
Azadirachta indica extract	none
Coscinium fenestratum extract	none
Curcuma longa, rhizoma pecodex extract	none
Note: RSD is < 7%	

A green tea extract and an oolong tea extract showed significant inhibition of adsorbed GTF enzyme. These extracts were progressed to evaluation in a rat caries study. Twice a day topical application of treatments resulted in less than a 15% reduction in caries.

CONCLUSION

Since plaque accumulation is thought to occur largely through replication of adhered bacteria versus continual deposition of bacteria, a GTF inhibitor may need to be present continuously and/or in combination with a growth inhibitor to be effective. Attempts at increasing substantivity of rat caries test solutions did not increase efficacy in this case.

Research presented at the 75th General Session of the IADR; Orlando, Florida March 19-23, 1997

P&G