Demonstration of the Robustness of a Well-established De/Remineralization Model

A. M. Pfarrer, R. A. Baker, C. M. McQueen and M. A. Kaminski*

P&G, Mason, OH, USA

2037

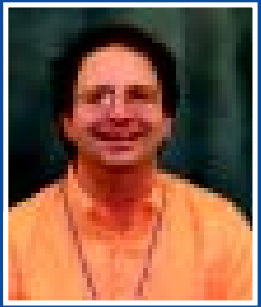

ABSTRACT

Many laboratory performance tests (LPT) used to screen the anticaries potential of fluoride-containing dentifrice formulations exist at single study sites and lack corroboration by separate researchers/sites. The acceptance and value of a test method lies in its ability to be duplicated in multiple laboratories. **Objective:** The objective of the following study was to assess the robustness of a well-established and validated pH cycling model, previously reported by Featherstone et al., by duplicating its response to fluoride at a separate oral biology testing facility. **Methods:** Caries-free human molar crowns (n=10) were cleaned and polished then treated twice/day with dentifrice slurries (1:3 slurry with water) for one minute. Between treatments, crowns were demineralized for 6hr (pH 4.4, Ca/P/acetate solution) and remineralized overnight (17 hr, pH 7, Ca/P). NaF with silica dentifrice formulations tested were: a) 0ppm F, b) 250ppm F, c) 1100ppm F and d) whitening toothpaste. After pH cycling, tooth samples were evaluated using cross-sectional microhardness. Delta Z values were calculated using microhardness indents. Results: Mean ± SE Delta Z values (vol % min x µm) were: a) 7154 ± 287, b) 3446 ± 198, c) 2396 ± 183 and d) 1521 ± 253, with d > c > b > a , ANOVA. **Conclusion: Although the mean Delta Z values are higher than what is usually seen the response to fluoride dose was excellent. The whitening dentifrice also exhibited performance similar to that previously reported when tested at the study designer's laboratory. These results demonstrate that the well-established model is robust and able to be duplicated at other oral biology research facilities.**

BACKGROUND

As required by law under the anticaries monograph, all fluoridated dentifrices must pass the Rat Caries Reduction model to demonstrate efficacy. In October of 2001, the FDA published a Request for Data on the use of intra oral-appliance models for compliance with biological testing requirements outlined in the anticaries monograph, i.e. rat caries model. In responding to this call for data, P&G proposed a well-characterized and validated *in vitro* model for the FDA to consider as an appropriate alternative. Additionally, we outlined the key elements to be considered when attempting to demonstrate equivalent accuracy.

- Provide a meaningful representation of the caries process with some correlation to clinical results.
- Demonstrate a dose response to fluoride with a clear separation in fluoride levels that have previously been shown to provide different levels of anticaries efficacy in the animal caries model.
- Clinically proven formulations perform similarly relative to the controls in both the pH cycling model and the animal caries model.
- Formulations previously shown to have attenuated fluoride activity in the animal caries model should be discernible in the alternative

MATERIALS AND METHODS

In an effort to replicate a well-established *in vitro* model in our laboratory and therefore demonstrate its robustness, caries-free, human crowns of molars and pre-molars were removed from the roots, brushed with warm detergent solution, polished with a 5 µm alumina slurry using a felt wheel to follow the contour of the tooth surface, rinsed in deionized water (DDW), air dried, and painted with acid resistant varnish leaving one exposed window (approximately 3.0 x 2.0 mm). The teeth were then suspended so that the windows were exposed to the test slurries at all times during the treatments and subsequent incubations in de- and remineralizing solutions. The treatments and incubations in de- and remineralization solutions, i.e., pH cycling, were repeated daily for a total of 14 days.

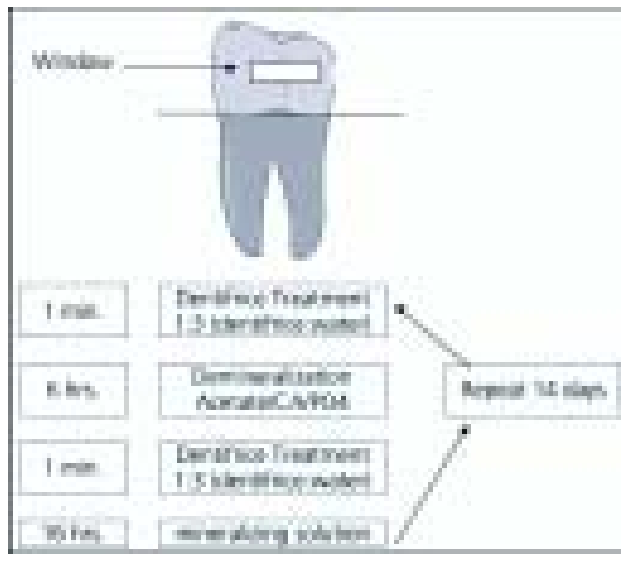
Window
1 min.
6 hrs.
Repeat 14 days

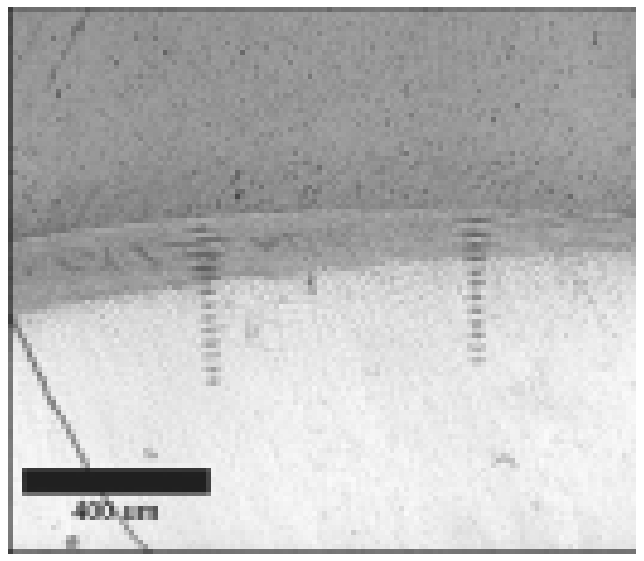
400 µm

After cycling, specimens were cut in half (through the lesion) using a radial saw. One half of each crown was embedded in epoxy resin so that the cut section of the lesion and underlying sound enamel were exposed. The cut surface was polished to a high luster and microhardness indentations were made on a line perpendicular to, and initiated at 12.5µm from the anatomical surface of the lesion. The hardness indenting was continued to depths extending into the underlying sound enamel, at 25µm intervals. Measurements were made using a standard microhardness indenter utilizing a Knoop diamond under a15g load for the outer 25µm measurements and a 50g load for all subsequent measurements). Knoop hardness numbers were subsequently converted to volume % mineral and individual volume % mineral values plotted versus depth permitting calculation of mineral loss (Delta Z).

RESULTS

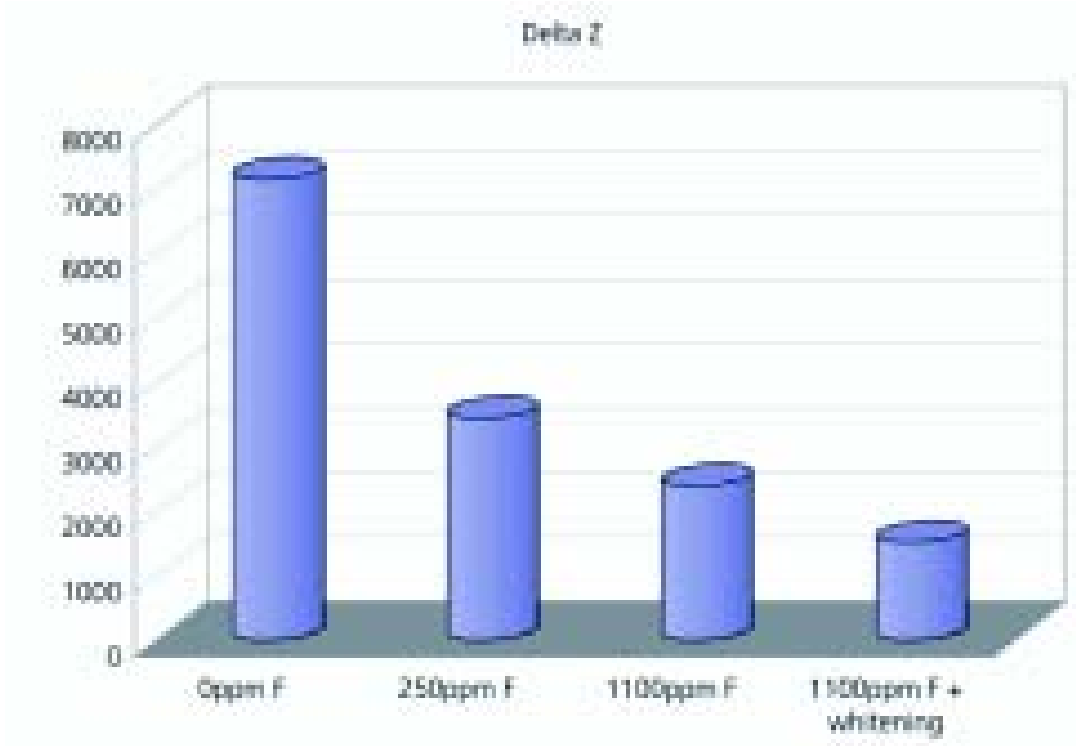
Delta Z
8000
7000
6000
5000
4000
3000
2000
1000
0
0ppm F
250ppm F
1100ppm F
1100ppm F + whitening

These results demonstrate that this well-establish and validated *in vitro* model can be reproduced in an independent laboratory. As expected, the model is sensitive to fluoride dose ($p < 0.05$). The relative performance of the test formulation replicated the results previously reported by Pfarrer et al., 2002. The greater Delta Z values observed in our laboratory are believed to be a result of removing more fluoride rich enamel during the preparation of the samples. Similar values have been reported by Jensen et al. at these proceedings in the same model using bovine enamel as a surrogate for human enamel.

CONCLUSION

- This *in vitro* pH cycling model was able to be performed in an alternate laboratory with minimal training.
- The results previously published were replicated in an alternate laboratory for both fluoride dose response formulations and the test formulation

References:
- *Pfarrer AM, White DJ, Rapozo-Hilo BS and Featherstone JDB. Anticaries and Hard Tissue Abrasion Effects of a "Dual-Action" Whitening, Sodium Hexametaphosphate Tartar Control Dentifrice. J CLin Dent 200213:50-54*
- *Jensen A, Toda S, Rapozo-Hilo M and Featherstone JDB. Development of an in vitro Bovine Enamel H-cycling Model. J Dent Res 84(Spec Iss A):#2548, 2005*
- *A comprehensive list of references describing this in vitro pH cycling model can be found in Pfarrer et al., 2002.*

MARCH 9-12, 2005

P&G

Research presented at the 83rd General Session of the IADR, March 9-12, 2005